NUC-3373 in combination with irinotecan (NUFIRI) or oxaliplatin (NUFOX) & bevacizumab for second-line treatment of patients with advanced colorectal cancer (NuTide:302)

Khurum Khan[1], Uma Mukherjee[1], Benjamin L Schlechter[2], Kristen K Ciombor[3], Aglaia Skolariki[4], Andrew L Coveler[5], Janet S Graham[6], Mary Linton Peters[7], Stacey A Cohen[5], Niharika B Mettu[8], Hélène Marijon[9], David Cosgrove[10], Fiona G McKissock[11], Elisabeth Oelmann[11], Jeffrey D Bloss[11], Sarah P Blagden[4], Aimery de Gramont[9], Jordan Berlin[3] and TR Jeffry Evans[6]

1) University College London NHS Foundation Trust, London, UK 2) Dana-Faber Cancer Institute, Boston, MA, USA 3) Vanderbilt University Medical Center, Nashville, TN, USA 4) Early Phase Clinical Trials Unit, Churchill Hospital, University of Oxford, Oxford, UK 5) Fred Hutchinson Cancer Center/University of Washington, Seattle, WA, US 6) Beatson West of Scotland Cancer Centre/University of Glasgow, Glasgow, UK 7) Beth Israel Deaconess Medical Center, Boston, MA, US 8) Duke University Medical Center, Durham, NC, US 9) Franco-British Institute, Levallois-Perret, France 10) Division of Medical Oncology, Vancouver Cancer Center, Compass Oncology, Vancouver, USA 11) NuCana plc, Edinburgh, UK

Poster Number: B048 **Email:** khurum.khan1@nhs.net

BACKGROUND

- CRC 3rd most common cancer[1]
- Global incidence 1.9 million and deaths 935,000 annually[1]
- 5-FU remains the cornerstone of treatment for CRC, despite several limitations
 - Rapidly degraded by DPD[2]
 - Short plasma half-life (8-14 mins)[3] requires long infusions (46-hour)
 - Generation of FBAL (associated with hand-foot syndrome)
 - Generation of FUTP (associated with diarrhea, mucositis, myelosuppression)
 - Cell entry requires nucleobase transporters
 - Complex enzymatic activation

NUC-3373 overcomes key limitations associated with 5-FU

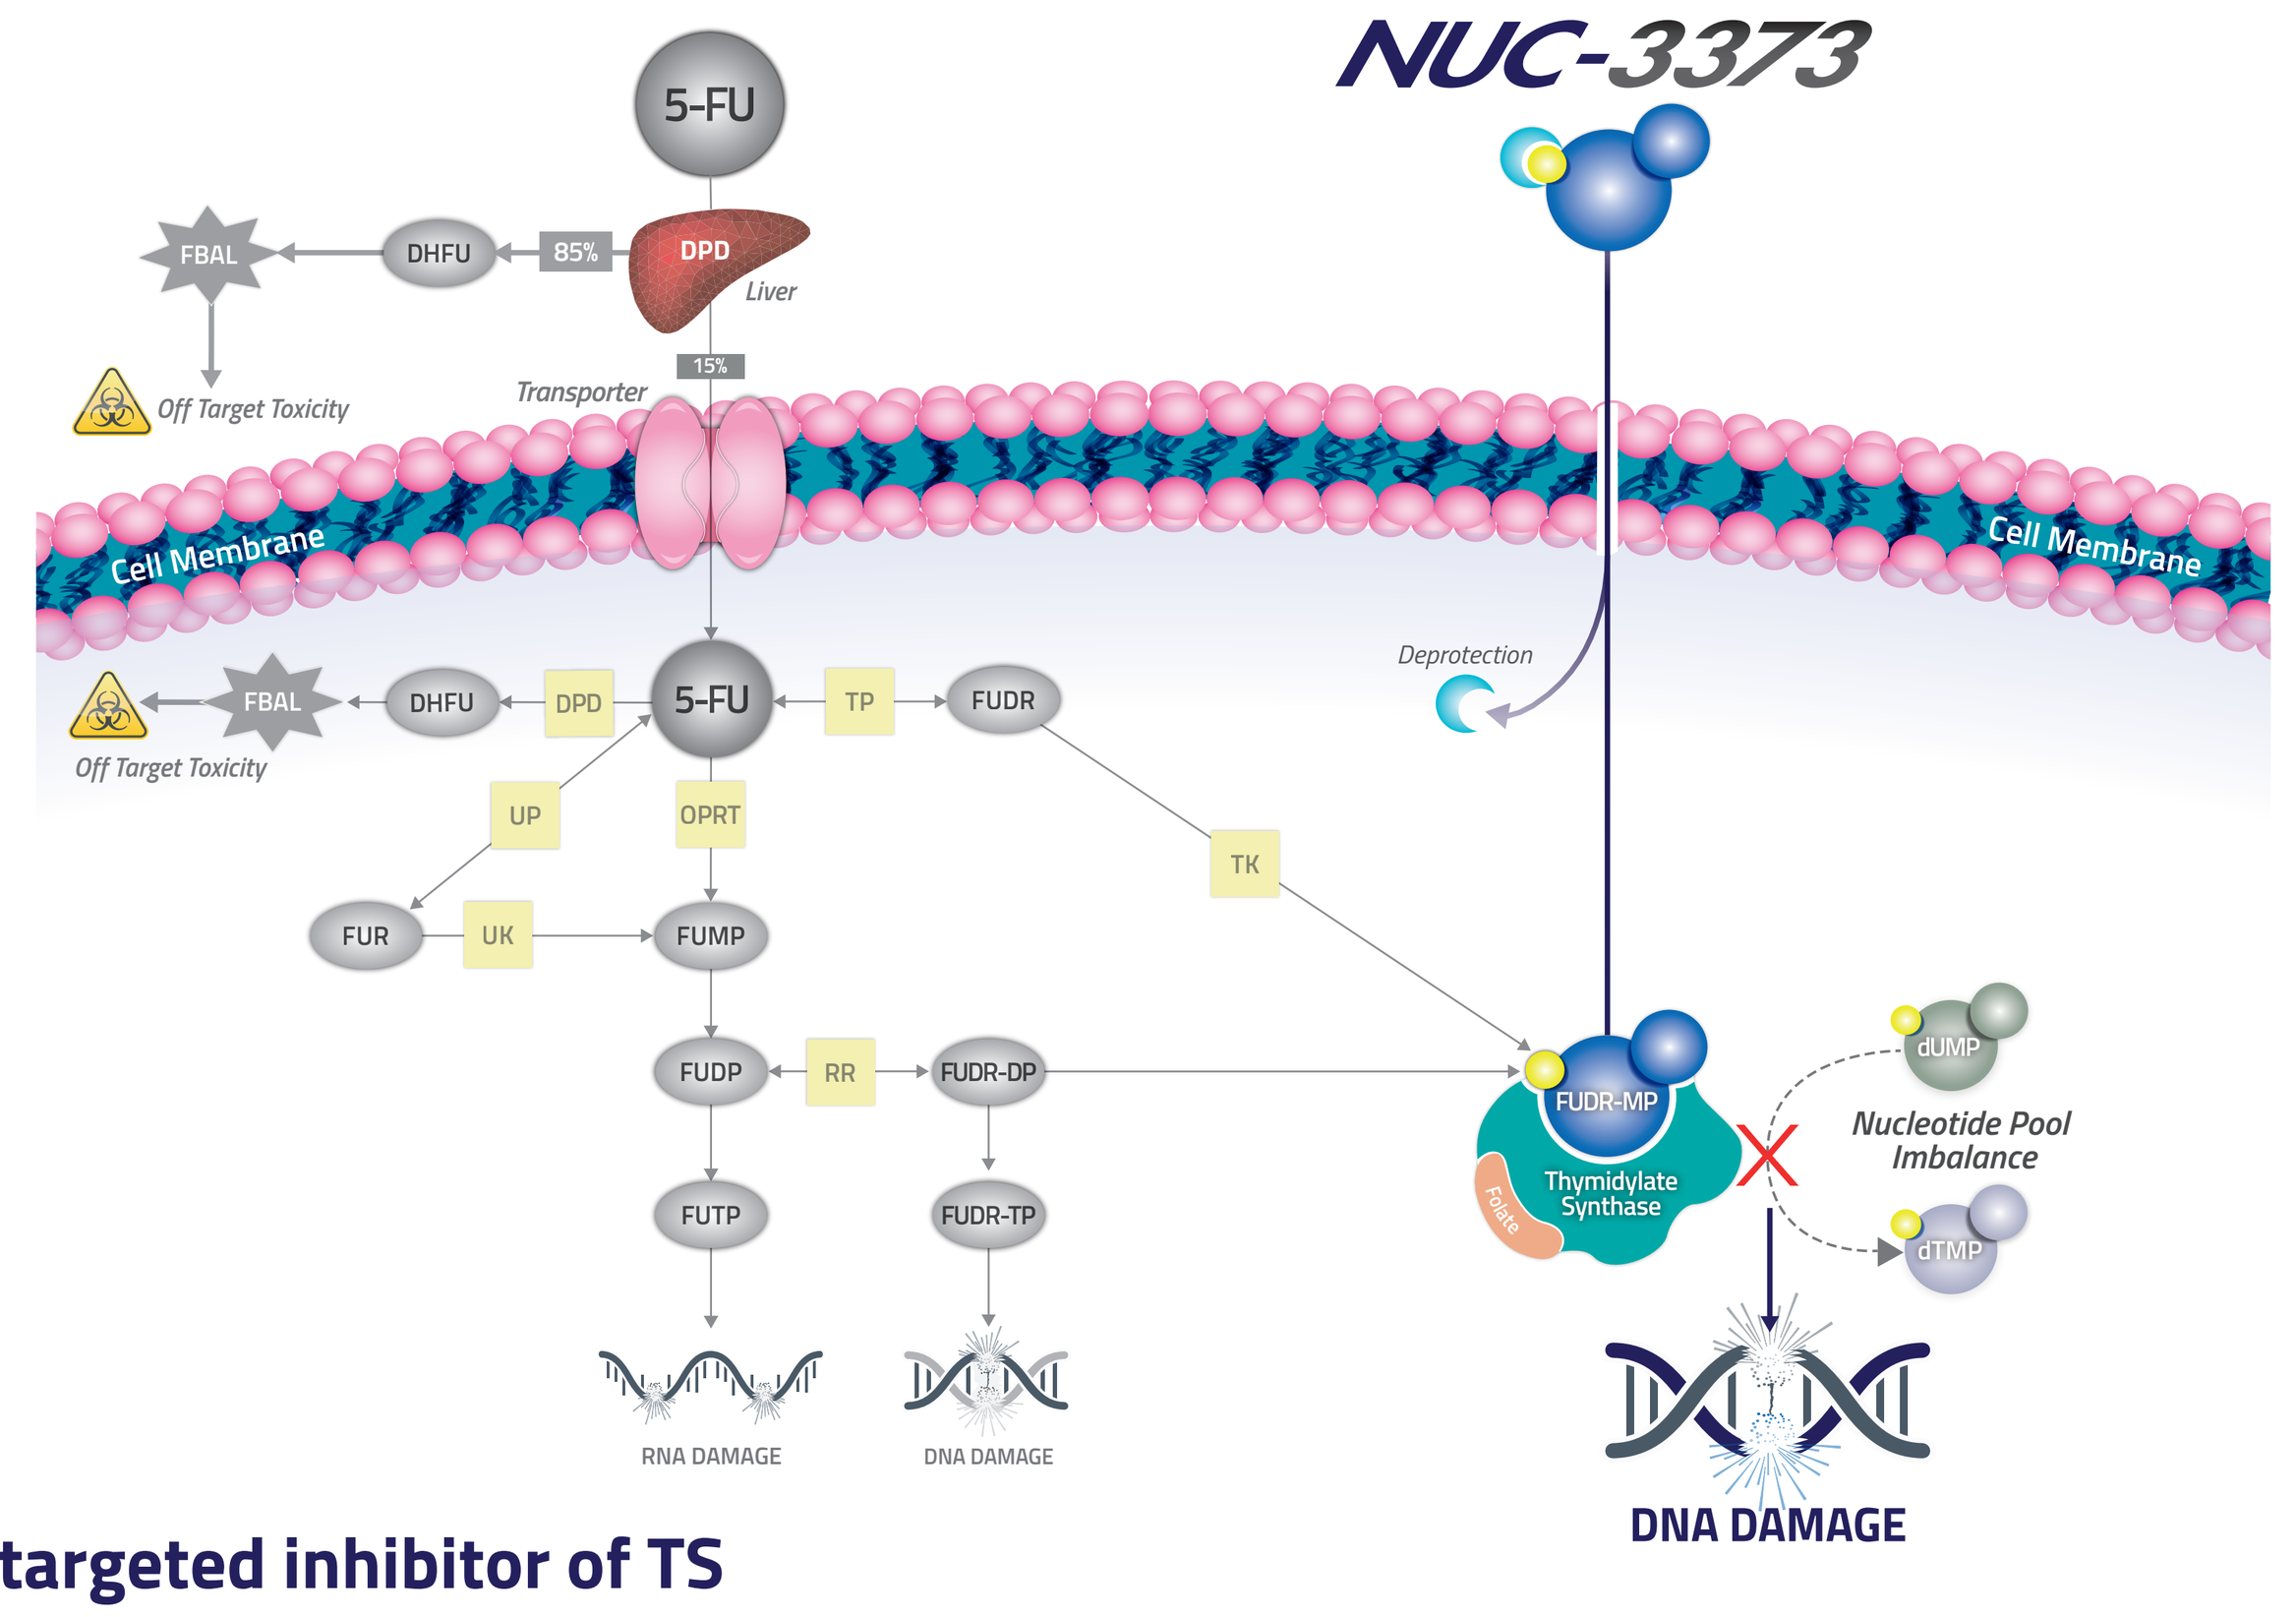
NUC-3373
5-FU
FBAL
DHFU
85%
DPD
Liver
15%
Off Target Toxicity
Transporter
Cell Membrane
Cell Membrane
Deprotection
FBAL
DHFU
DPD
5-FU
TP
FUDR
Off Target Toxicity
UP
OPRT
TK
FUR
UK
FUMP
FUDP
RR
FUDR-DP
FUDR-MP
Thymidylate Synthase
Folate
dUMP
Nucleotide Pool Imbalance
dTMP
FUTP
FUDR-TP
RNA DAMAGE
DNA DAMAGE
DNA DAMAGE

NUC-3373: A targeted inhibitor of TS

- Phosphoramidate transformation of FUDR[4,5,]
 - Resistant to breakdown by DPD
 - Enters cells independently of nucleobase transporters
 - Directly delivers FUDR-MP intracellularly
 - Low levels of toxic metabolites (FBAL, FUTP)
- Generates high intracellular levels of active anti-cancer metabolite FUDR-MP[6]
 - Causes an imbalance in the nucleotide pool leading to DNA damage and cell death[6]
 - Induces ER stress and DAMPs release leading to immunogenic cell death[7,8]
- Long plasma half-life allows for short infusion duration

NuTide:302 Study - Phase 1b/2

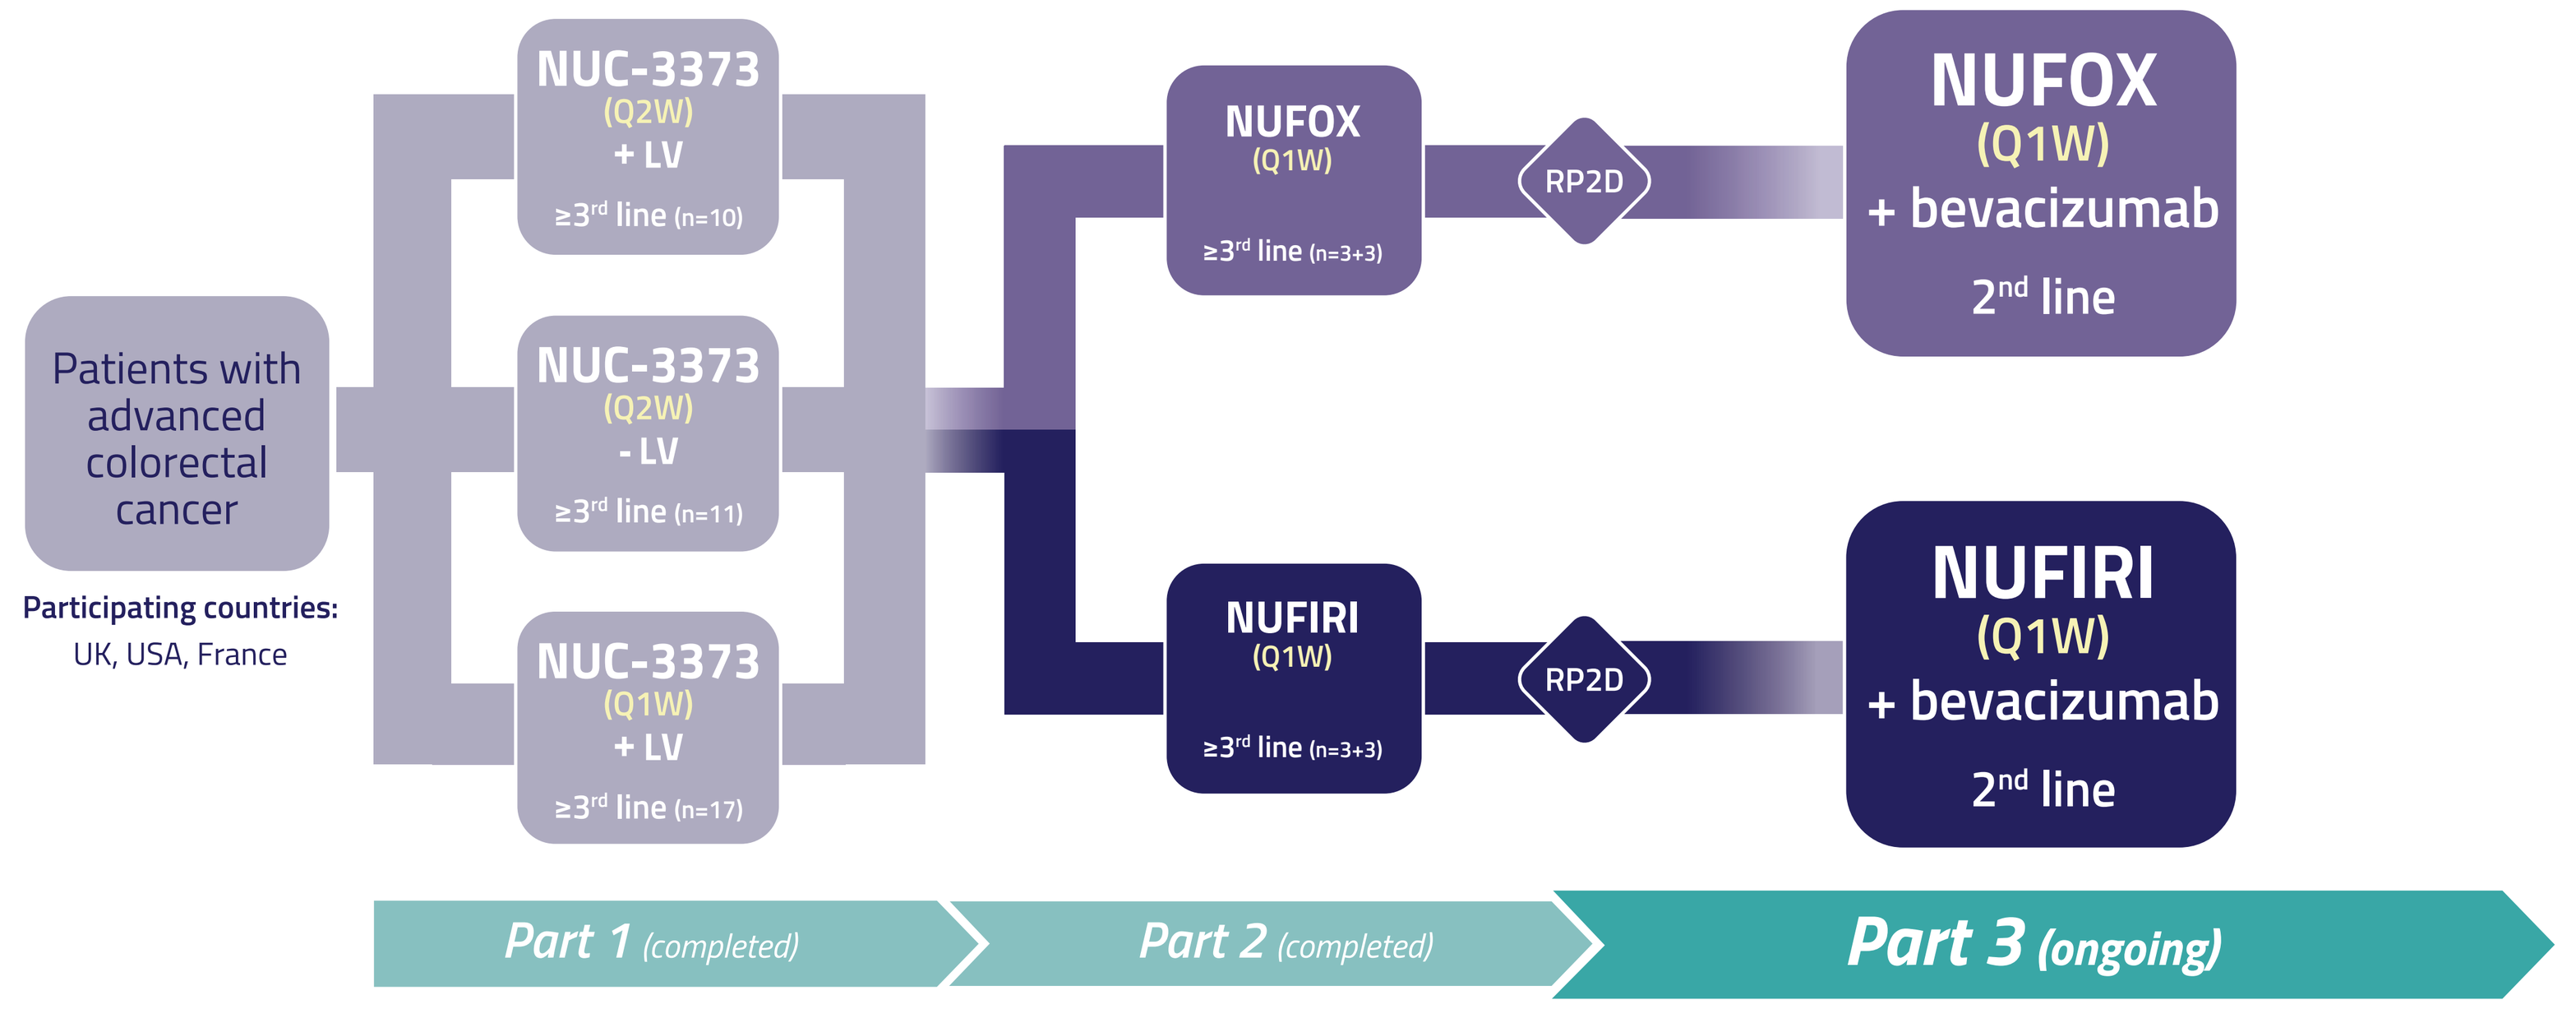
Patients with advanced colorectal cancer
Participating countries: UK, USA, France
NUC-3373 (Q2W) + LV ≥3rd line (n=10)
NUC-3373 (Q2W) - LV ≥3rd line (n=11)
NUC-3373 (Q1W) + LV ≥3rd line (n=17)
NUFOX (Q1W) ≥3rd line (n=3+3)
NUFIRI (Q1W) ≥3rd line (n=3+3)
RP2D
RP2D
NUFOX (Q1W) + bevacizumab 2nd line
NUFIRI (Q1W) + bevacizumab 2nd line
Part 1 (completed)
Part 2 (completed)
Part 3 (ongoing)

NUFOX-bev = 1,875 mg/m² NUC-3373 (Q1W), 400 mg/m² LV (Q1W), 85 mg/m² oxaliplatin (Q2W) and 5 mg/kg bevacizumab (Q2W)

NUFIRI-bev = 1,500 mg/m² NUC-3373 (Q1W), 400 mg/m² LV (Q1W), 180 mg/m² irinotecan (Q2W) and 5 mg/kg bevacizumab (Q2W)

Objectives: Anti-cancer activity, safety, PK of NUC-3373 in combination with oxaliplatin and bevacizumab or irinotecan and bevacizumab

NUFOX-bev

Patients with mCRC who had received 1 prior fluoropyrimidine and irinotecan-containing therapy

Baseline Characteristics (n=6)

Age, years	median (range)	64 (37-72)
Sex	male	3 (50%)
	female	3 (50%)
ECOG PS	0	0
	1	6 (100%)
Metastatic sites	≤3	1 (17%)
	≥4	5 (83%)
Liver metastases	Y	3 (50%)
	N	3 (50%)
BRAF status	wt	5 (83%)
	mt	0
	unknown	1 (17%)
RAS status	wt	3 (50%)
	mt	3 (50%)
Prior Bev	Y	0
	N	6 (100%)
Adjuvant CT	Y	5 (83%)
	N	1 (17%)

Treatment Related AEs (n=6)

	All Grades	Grade 3
Nausea	6 (100%)	1 (17%)
Diarrhea	5 (83%)	0
Fatigue	3 (50%)	0
Flushing	3 (50%)	0
Vomiting	3 (50%)	1 (17%)
Headache	3 (50%)	0
Abdominal pain	2 (33%)	0
Constipation	2 (33%)	0
Decreased appetite	2 (33%)	0
Dizziness	2 (33%)	0
Anemia	1 (17%)	0
Dysgeusia	1 (17%)	0
Platelet count decreased	1 (17%)	0

All Grade TRAEs with an incidence of ≥10% in combined NUFIRI/NUFOX population. NUC-3373 ± combinations related AEs

- No Grade 4 or 5 TRAEs
- 2 pts experienced Grade 3 TRAEs; nausea & vomiting (1), hypokalemia (1)
- No discontinuations due to AEs
- NUFOX-bev well-tolerated

Progression Free Survival

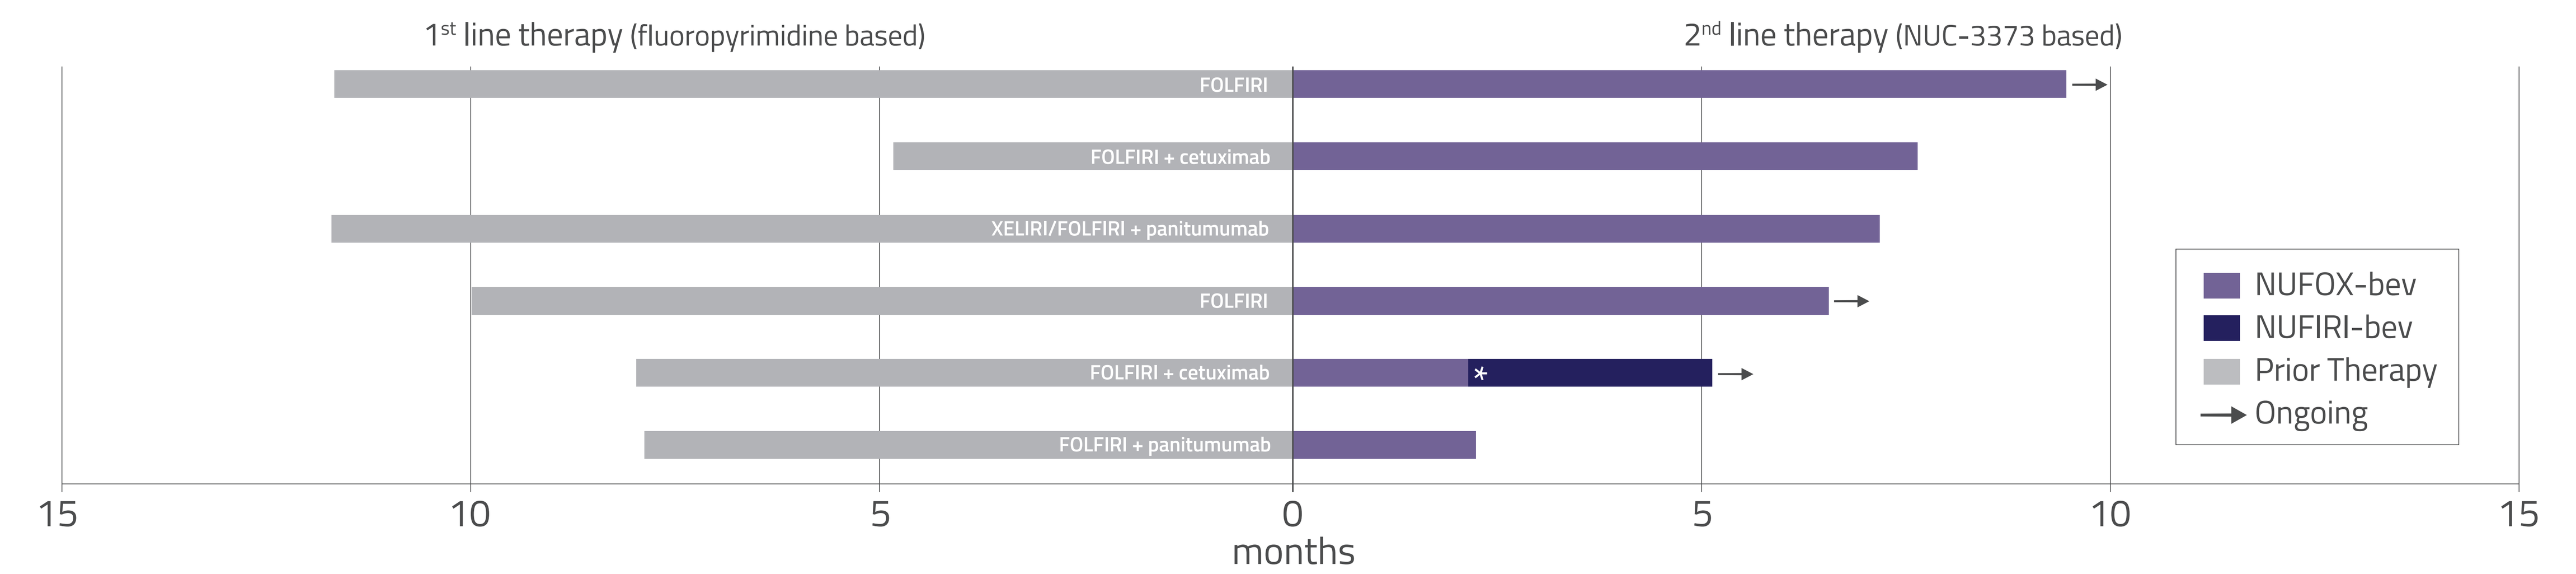
1st line therapy (fluoropyrimidine based)
2nd line therapy (NUC-3373 based)
FOLFIRI
FOLFIRI + cetuximab
XELIRI/FOLFIRI + panitumumab
FOLFIRI
FOLFIRI + cetuximab
FOLFIRI + panitumumab
*
NUFOX-bev
NUFIRI-bev
Prior Therapy
Ongoing
15
10
5
0
5
10
15
months

Standard fluoropyrimidine based CRC regimens ~5 months longer PFS in 1st vs 2nd line

*switched to NUFIRI-bev due to oxaliplatin-related infusion reaction

Best % Change in Tumor Burden

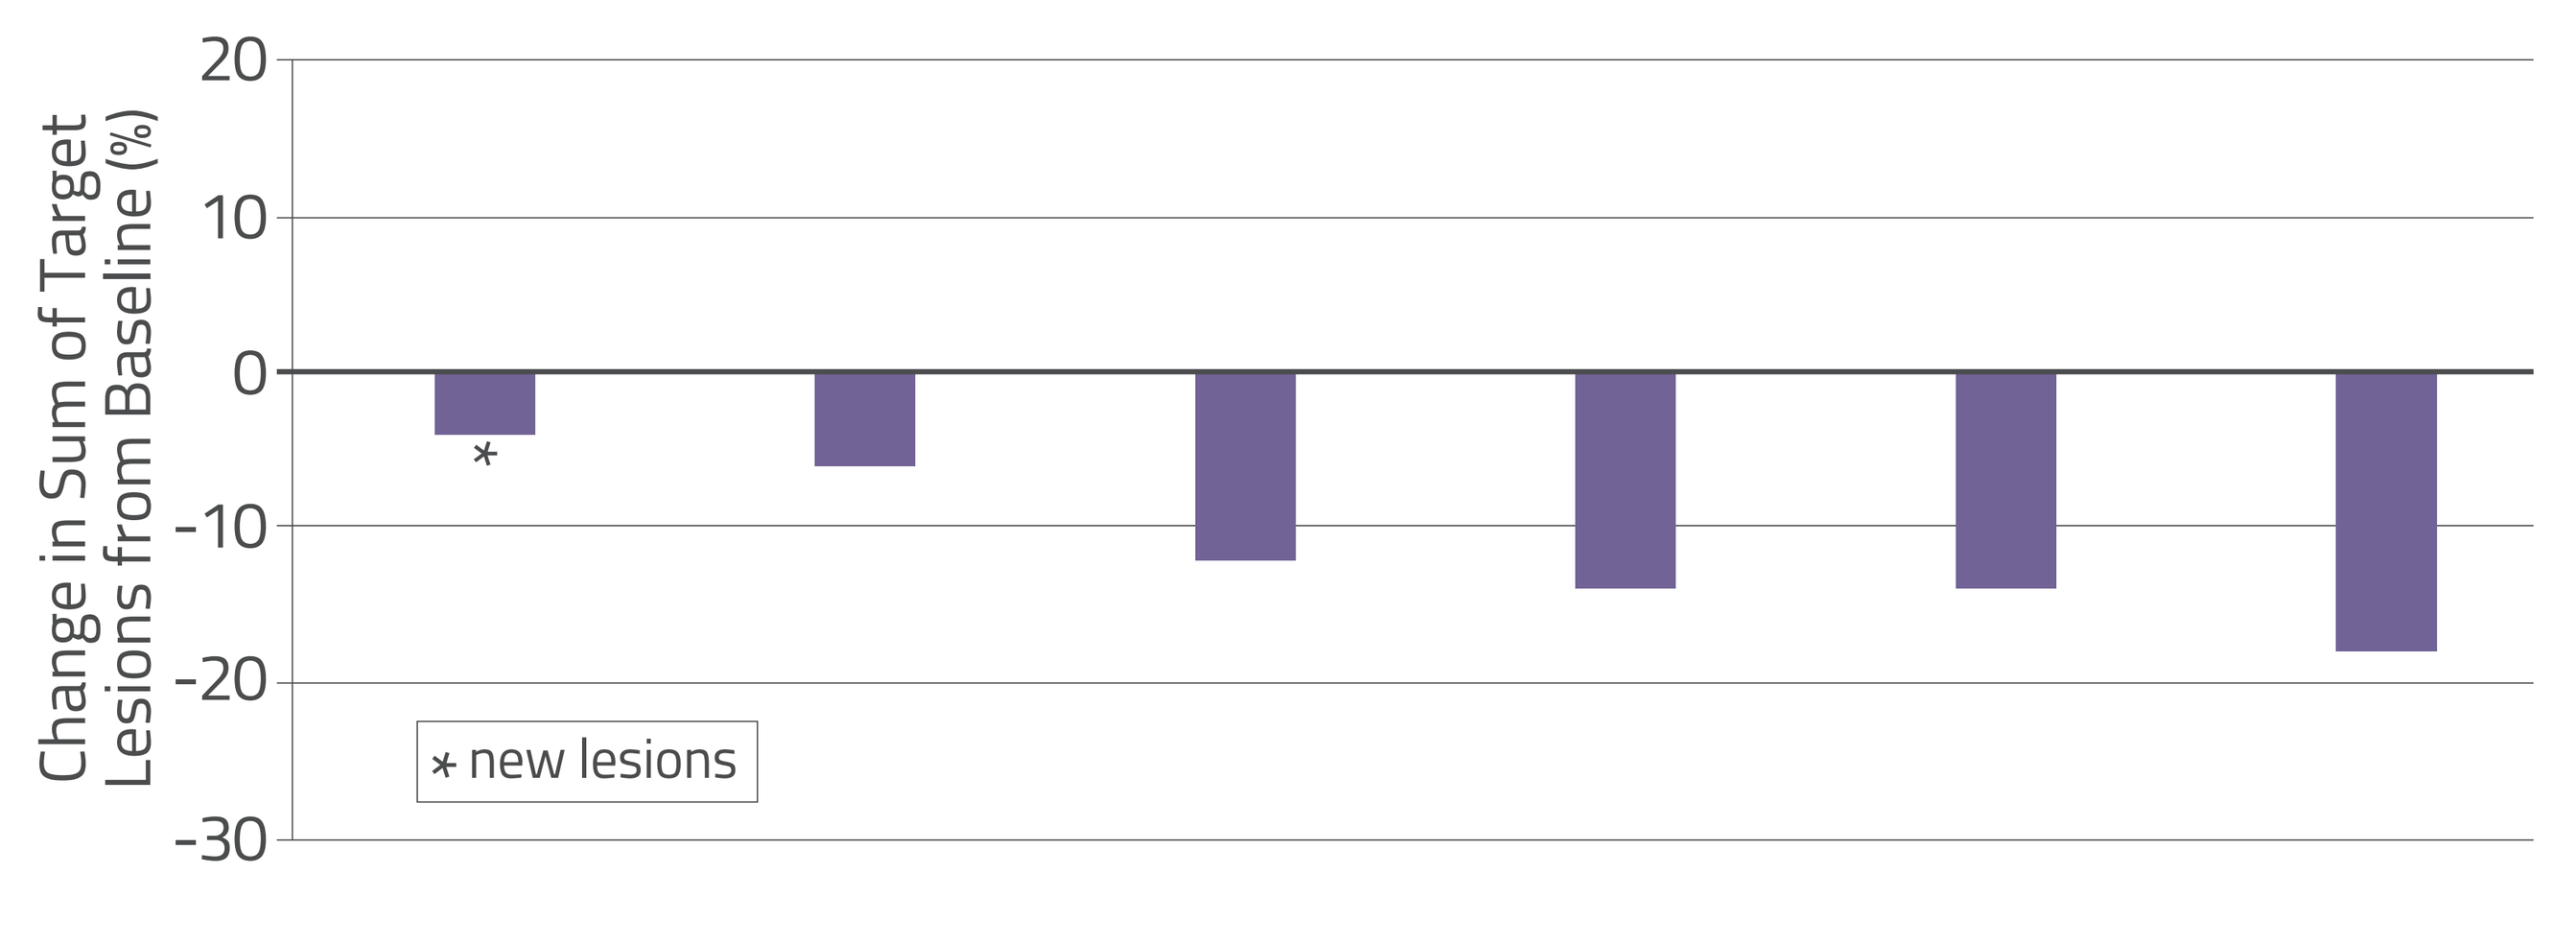
Change in Sum of Target Lesions from Baseline (%)
20
10
0
-10
-20
-30
*
* new lesions

NUFIRI-bev

Patients with mCRC who had received 1 prior fluoropyrimidine and oxaliplatin-containing therapy

Baseline Characteristics (n=8)

Age, years	median (range)	56 (40-81)
Sex	male	6 (75%)
	female	2 (25%)
ECOG PS	0	5 (63%)
	1	3 (38%)
Metastatic sites	≤3	1 (13%)
	≥4	7 (88%)
Liver metastases	Y	4 (50%)
	N	4 (50%)
BRAF status	wt	6 (75%)
	mt	1 (13%)*
	unknown	1 (13%)
RAS status	wt	4 (50%)
	mt	4 (50%)
Prior Bev	Y	1 (13%)
	N	7 (88%)
Adjuvant CT	Y	1 (13%)
	N	7 (88%)

*3rd line patient

Treatment Related AEs (n=8)

	All Grades	Grade 3
ALT increased	5 (63%)	2 (25%)
AST increased	5 (63%)	0
Diarrhea	5 (63%)	0
Nausea	4 (50%)	0
Anemia	3 (38%)	0
Fatigue	2 (25%)	0
Flushing	2 (25%)	0
Vomiting	2 (25%)	0
Abdominal pain	1 (13%)	0
Constipation	1 (13%)	0
Decreased appetite	1 (13%)	0
Dysgeusia	1 (13%)	0
Platelet count decreased	1 (13%)	0

All Grade TRAEs with an incidence of ≥10% in combined NUFIRI/NUFOX population. NUC-3373 ± combinations related AEs

- No Grade 4 or 5 TRAEs
- 2 pts experienced Grade 3 TRAEs; ALT increased & pancreatitis (1), ALT increased (1)
- No discontinuations due to AEs
- NUFIRI-bev well-tolerated

Progression Free Survival

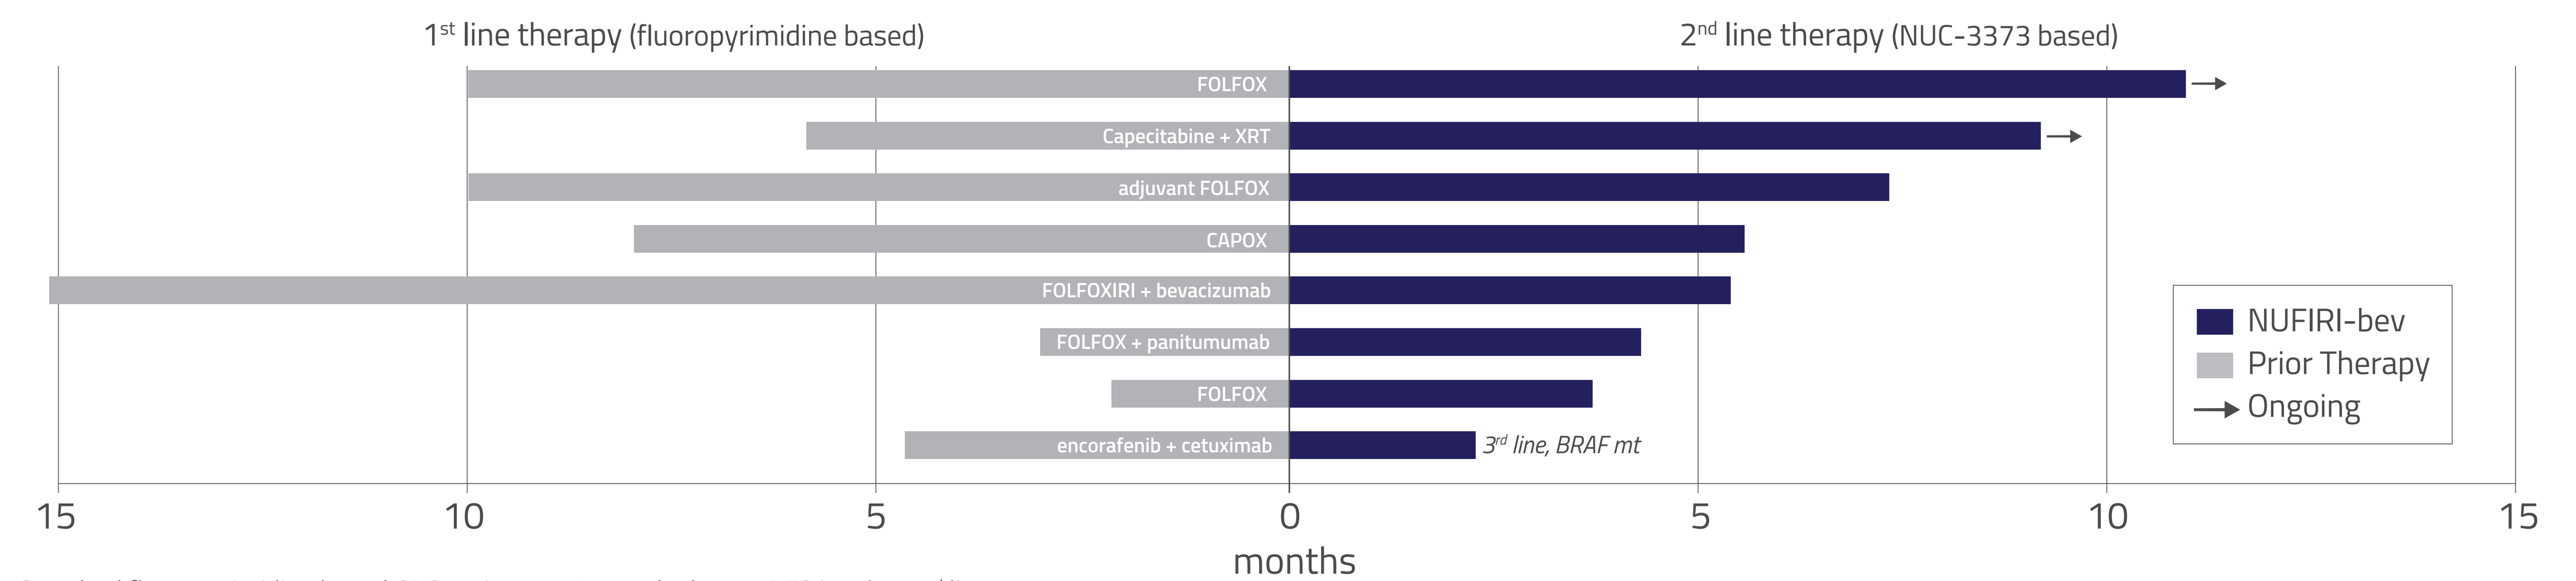
1st line therapy (fluoropyrimidine based)
2nd line therapy (NUC-3373 based)
FOLFOX
Capecitabine + XRT
adjuvant FOLFOX
CAPOX
FOLFOXIRI + bevacizumab
FOLFOX + panitumumab
FOLFOX
encorafenib + cetuximab
3rd line, BRAF mt
NUFIRI-bev
Prior Therapy
Ongoing
15
10
5
0
5
10
15
months

Standard fluoropyrimidine based CRC regimens ~5 months longer PFS in 1st vs 2nd line

CONCLUSION

- NUC-3373 can be combined with agents commonly used in CRC at their standard doses
- Favorable safety profiles for NUFIRI-bev & NUFOX-bev
- Prolonged disease control and signs of anti-tumor activity
 - Several patients achieved similar or greater PFS in 2nd line NUC-3373 based therapy vs 1st line fluoropyrimidine based therapy
- More convenient dosing schedule compared to 5-FU (2-2.5h vs 46h)
- Randomized Phase 2 study currently investigating safety and efficacy of NUFIRI-bev vs FOLFIRI-bev in 2nd line patients with CRC (NuTide:323, NCT05678257)

Best % Change in Tumor Burden

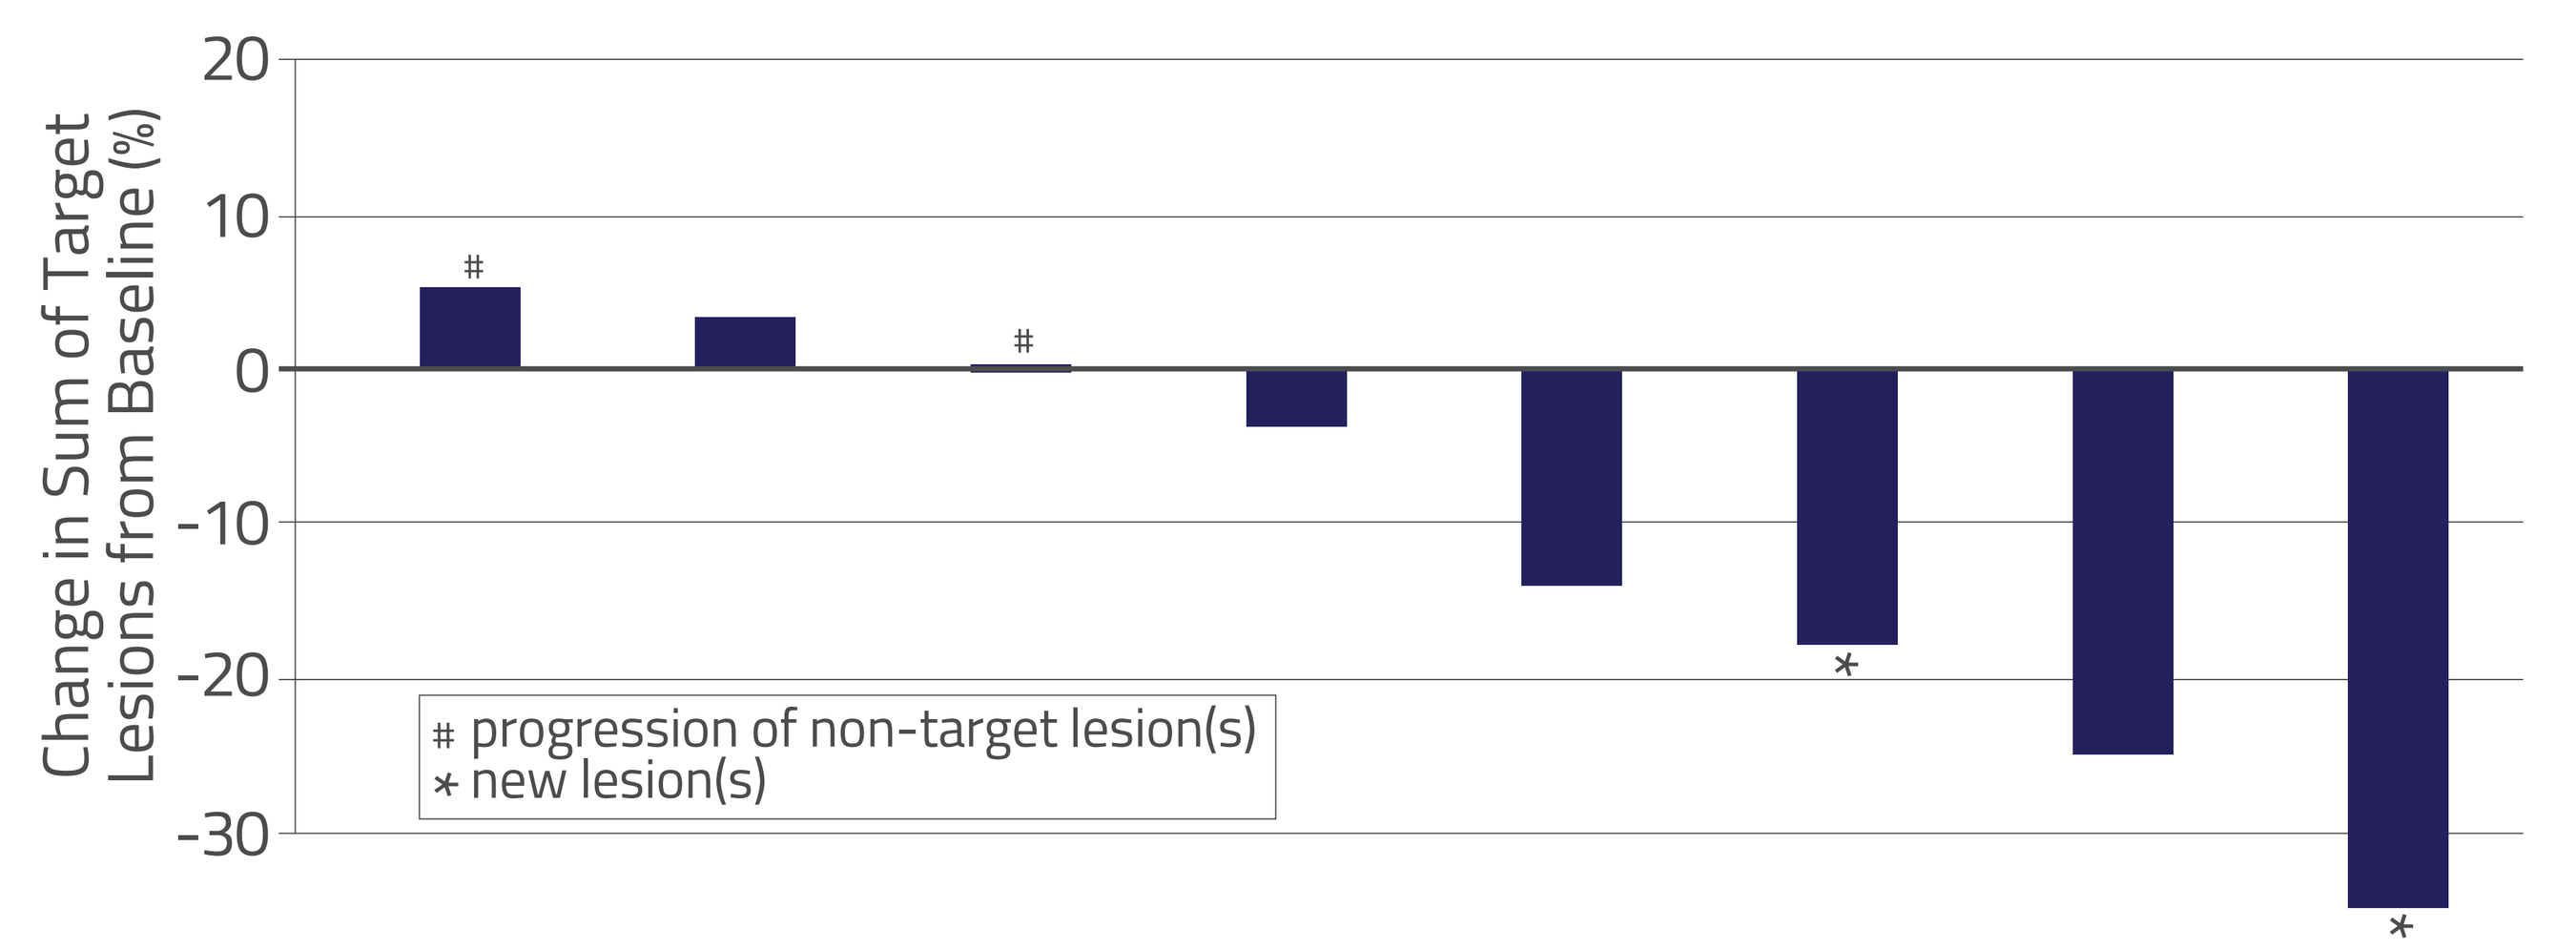
Change in Sum of Target Lesions from Baseline (%)
20
10
0
-10
-20
-30
#
#
*
*
progression of non-target lesion(s)
* new lesion(s)

REFERENCES: 1. World Health Organization International Agency for Research on Cancer (IARC). GLOBOCAN 2020: Available from :https:/gco.iarc.fr/today/data/factsheets/cancers/10_8_9-Colorectum-fact-sheet.pdf 2. Diasio & Harris, 1999 *Clin Pharmacokinet*; 16:215-237 3. Heggie *et al*, 1987 *Cancer Research*; 47:2203-2206 4. McGuigan *et al*, 2011 *Med Chem*; 27: 7247-7258 5. Vande Voorde *et al*, 2011 *Biochem Pharmacol*; 82: 441-452 6. Bre *et al*, 2023 *Cancer Chem Pharm*; 91(5):401-412 7. McKissock *et al*, 2020 *Cancer Res*; 80: Suppl_16:1848 8. Read *et al*, 2022 *Cancer Res*; 82:Suppl_12:1113-1113

ABBREVIATIONS: 5-FU: 5-fluorouracil AE: adverse event ALT: alanine aminotransferase Bev: bevacizumab BRAF: v-raf murine sarcoma viral oncogene homolog B1 CAPOX: capecitabine + oxaliplatin CT: chemotherapy CRC: colorectal cancer DAMP: damage-associated molecular pattern DHFU: dihydrofluorouracil DNA: deoxyribonucleic acid DPD: dihydropyrimidine dehydrogenase dUMP: deoxyuridine monophosphate dTMP: deoxythymidine monophosphate ECOG PS: Eastern Cooperative Oncology Group performance score ER: endoplasmic reticulum FBAL: alpha-fluoro-beta-alanine FOLFIRI: 5-FU + LV + irinotecan FOLFOX: 5-FU + LV + oxaliplatin FUDP: fluorouridine diphosphate FUDR: fluorodeoxyuridine FUDR-DP: fluorodeoxyuridine diphosphate FUDR-MP: fluorodeoxyuridine monophosphate FUDR-TP: fluorodeoxyuridine triphosphate FUTP: fluorouracil triphosphate LV: leucovorin mCRC: metastatic colorectal cancer mt: mutant NUFIRI: NUC-3373 + LV + irinotecan NUFOX: NUC-3373 + LV + oxaliplatin OPRT: orotate phosphoribosyl transferase PFS: progression free survival PK: pharmacokinetics Q1W: once weekly Q2W: once every 2 weeks RP2D: recommended phase 2 dose RR: ribonucleotide reductase TK: thymidine kinase TP: thymidine phosphorylase TRAE: treatment related adverse event TS: thymidylate synthase UK: uridine kinase UP: uridine phosphorylase wt: wild type XELIRI: irinotecan and capecitabine XRT: radiotherapy

Copies of this poster obtained through QR (Quick Response) and/or text key codes are for personal use only and may not be reproduced without written permission of the authors.

Data cleaning ongoing: data cut-off date 22 August 2023